DEVELOPMENT OF « READY TO USE » INFUSIONS OF BUPIVACAINE, FENTANYL AND MORPHINE FOR USE IN ANAESTHESIOLOGY

AN INTERDISCIPLINARY APPROACH

W.Griffiths [1] , B. Kramer [2] , T. Nguyen-Xuan [2] , C. Kern [3] , E. Van Gessel [3] , P. Bonnabry [1].

[1] Pharmacy, [3] Department of Anaesthesiology, University Hospitals of Geneva (HUG)

[2] Bioren SA, Couvet, Switzerland

Background and Objective

Medical errors are one of the leading causes of death in the hospital environment[1]. Amongst them, drug administration is in the forefront[2]: analgesics being the primary drugs involved[3]. When using patient-controlled analgesia (PCA) devices for post operative pain management, several types of drugs are administered: local anaesthetics (LA) such as bupivacaine for continuous nerve block, LA with opiates for epidural injection or morphine for intravenous analgesia. Bupivacaine is a standard in obstetric epidural block or post-operative analgesia and it has been shown that the addition of a liposoluble opiate such as fentanyl at 2 mcg/ml improves analgesic quality[4]. Until recently these complex solutions were prepared extemporaneously, either by anaesthesiologists or nursing staff in the operating theatre or recovery room, or by pharmacy-based centralised intravenous additive (CIVA) services using aseptic techniques. The aim of this study was to develop ready-to-use solutions of morphine as well as bupivacaine with fentanyl at an industrial level using steam sterilisation if possible, and with a long shelf life.

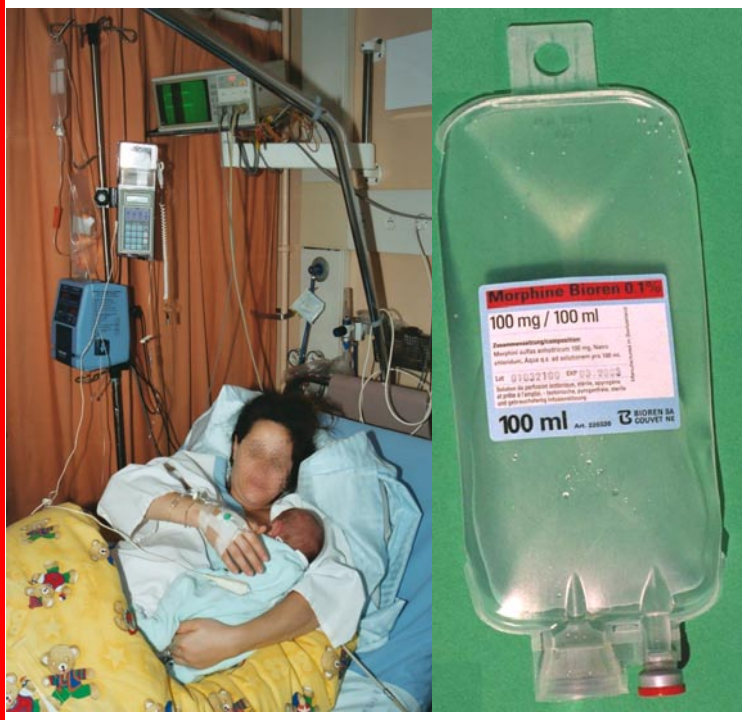
Morphine Bioren 0.1%
100 mg / 100 ml
100 ml

IV PCA after caesarian section under spinal anaesthesia

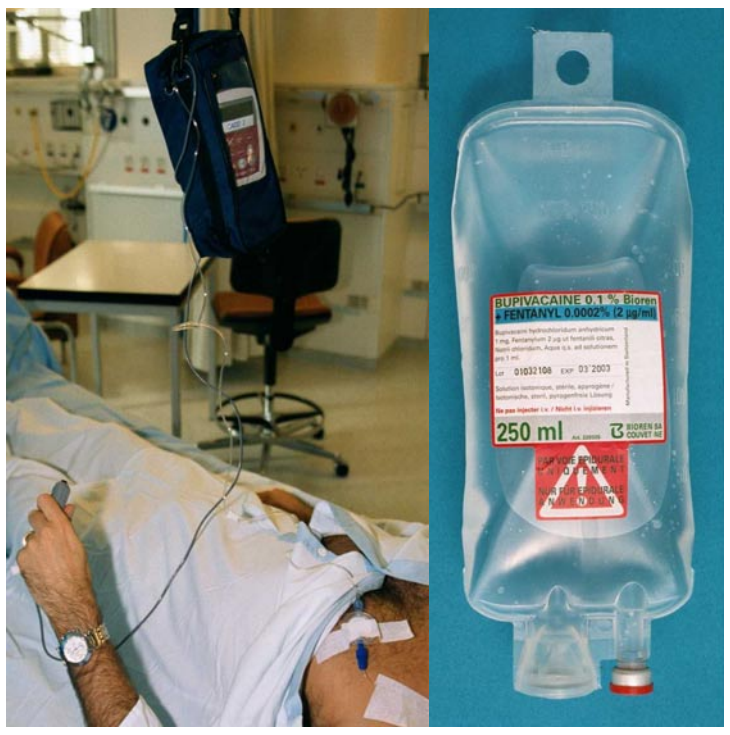
BUPIVACAINE 0.1 % Bioren
+ FENTANYL 0.0002% (2 µg/ml)
250 ml

Epidural PCA in the recovery room

William Griffiths

Methods

Discussions between the Pharmacy, the Department of Anaesthesiology HUG and a Swiss pharmaceutical company led to the manufacture of a morphine solution 100mg/100ml for PCA, and bupivacaine 0.125% with fentanyl 0.0002% (250ml) in polypropylene infusion bags (Bioren) for use in both general and obstetric surgery. The bags were filled with the solutions at 80°C using a semi-automatic filling machine and sterilised at 121°C for 20 minutes. Stability tests were then carried out at room temperature (RT): 25 ± 2 °C with a relative humidity (RH) of 60 ± 5%, an accelerated test at 30 ± 2 °C with an RH of 60 ± 5% (AT1) and another at 40 ± 2 °C with 75% ± 5% RH (AT2) using HPLC analytical methods.

Morphine 0.1% (100 ml)

HPLC Parameters
Hewlett Packard 1100

Column:	Nucleosil C18 (5 µm) 250 x 4.6 mm 25 °C
Detection:	UV 283 nm
Injection Vol.:	20 µl
Mobile phase:	Na Octane-sulfonate 0.148% aqueous solution : Acetonitrile : Acetic acid 73 : 25 : 2 1.5 ml/min.

Bupivacaine 0.125% - Fentanyl 0.0002% (250 ml)

HPLC Parameters
Hewlett Packard 1100

Column:	Inertsil 5 ODS2, 250 x 4.6 mm 30 °C
Detection:	UV 265 & 210 nm
Injection Vol.:	50 µl
Mobile phase:	Phosphate buffer 0.02 M pH 8.0 : Acetonitrile 40 : 60 1 ml/min.

Results

The results show that the finished products are stable after 9 months at RT and 6 months at AT1 and AT2. The ultimate goal is for a shelf life of 3 years. Both preparations will be submitted to the swiss health authorities for registration at the end of 2002.

MORPHINE 0.1% 100 ml		Stability N° 01002 Lot N° 01032100				Stability N° 01002 Lot N° 01032101			
Assays	Specifications	START	3 months	6 months	9 months	START	3 months	6 months	9 months
Storage condtions : RT									
Morphine	92.5 - 105.0 %	100.5	101.4	102.0	102.1	100.6	100.5	101.0	101.3
Storage condtions : AT1									
Morphine	92.5 - 105.0 %	100.5	101.5	102.3		100.6	100.6	101.4	
Storage condtions : AT2									
Morphine	92.5 - 105.0 %	100.5	100.8	100.9		100.6	99.9	101.6	

BUPIVACAINE 0.125 % FENTANYL 0.0002% 250 ml		Stability N° 01004 Lot N° 01032109				Stability N° 01005 Lot N° 01032110			
Assays	Specifications	START	3 months	6 months	9 months	START	3 months	6 months	9 months
Storage condtions : RT									
Bupivacaine hydrochloride	92.5 - 105.0 %	100.3	100.4	100.8	101.2	101.1	100.3	101.4	101.8
Fentanyl citrate	92.5 - 105.0 %	100.7	99.5	99.4	99.8	103.8	102.0	103.3	103.9
Storage condtions : AT1									
Bupivacaine hydrochloride	92.5 - 105.0 %	100.3	100.7	101.3		101.1	101.4	102.0	
Fentanyl citrate	92.5 - 105.0 %	100.7	99.5	99.4		103.8	103.3	103.6	
Storage condtions : AT2									
Bupivacaine hydrochloride	92.5 - 105.0 %	100.3	100.5	100.8		101.1	101.2	101.6	
Fentanyl citrate	92.5 - 105.0 %	100.7	99.0	99.0		103.8	103.3	103.0	

Conclusions

Ready-to-use solutions facilitate prescribing due to: standard doses, reduced manipulation, diminished potential errors (under- or overdose), ease of use, gain in time, low risk of contamination during preparation, significant cost savings and ultimately more prescriptions which is an advantage for the patient. These preparations have already been beneficial to 70% of the patients in confinement at the HUG maternity. In the field results, after several months use, showed that a return to traditional methods would be felt as a step backwards by both the anaesthesiologists and the nursing staff of the HUG. The collaboration between the departments of pharmacy and anaesthesiology and the pharmaceutical industry has resulted in stable solutions with improved quality and safety for epidural injection and post operative analgesia.

References

1. Building a Safer NHS for Patients. 2001. www.doh.gov.uk/buildsafenhs/
2. Cadman B. Hosp. Pharm. July 1999; 6 : 213 - 218.
3. Bates.DW. JAMA 1995; 274: 29 - 34
4. Lyons G. et al. Br J Anaesth 1997; 78: 493-6

HUG Hôpitaux Universitaires de Genève

EAHP Vienna 2002

BIOREN